Phase 2 Trial of Oncolytic Vaccinia Virus Olvi-Vec-Primed Immunochemotherapy In Heavily Pretreated Platinum-Resistant/Refractory Ovarian Cancer (PRROC)

Robert W. Holloway[a], Alberto A. Mendivil[b], James E. Kendrick[a], Lisa N. Abaid[b], John V. Brown[b], Christine K. Fitzsimmons[a], Jessica A. Kennard[a], Monica King[b], Jane LeBlanc[a], Katrina Lopez[b], Madhavi Manyam[a], Nathalie D. McKenzie[a], Kristina M. Mori[b], Jamie L. Smith[a], Amanda J. Stephens[a], Sarfraz Ahmad[a]

[a]AdventHealth Cancer Institute, Orlando, FL 32804; [b]Gynecologic Oncology Associates, Newport Beach, CA 92663, USA

Background and Rationale

❖ **Olvi-Vec (olvimulogene nanivacirepvec)**

- Modified oncolytic vaccinia virus (LIVP strain; aka GL-ONC1; laboratory name: GLV-1h68) with mutations that enhance tumor targeting (Zhang 2009)
- Olvi-Vec triggers oncolysis, immunogenic cell death, and augmented tumor (neo)antigen presentation (Worschech 2009)
- *In vitro* and *in vivo* synergy with platinum & other cytotoxics (Song 2007; Binz 2017)

❖ **Olvi-Vec turns 'Cold' ovarian tumor to 'Hot'**

- Activates innate immunity & inflammatory response (Worschech 2009)
- Enhances tumor-infiltrating lymphocytes (TILs) & adaptive immunity (see translational data)

❖ **Olvi-Vec re-sensitizes tumors to platinum-based therapy**

- Killing of chemoresistant cancer stem cells
- Enhanced intratumoral infiltration of CD8+ T cells changes glutathione & cysteine metabolism and abolish tumor stromal fibroblast-mediated platinum resistance (Wang 2016)
- Up-regulation of intratumoral STAT1 expression by Olvi-Vec; STAT1 is a predictive biomarker of response in high-grade serous ovarian carcinoma (Au 2016)
- Enhanced intratumoral inducible nitric oxide synthase (iNOS) expression; Favorable shift M2 → M1 iNOS$^+$ MDSC (Kilinc 2016)
- Chemotherapies reduce tumor immune suppression, enhance antigen presentation by ICD (Emens & Middleton 2015), and boost antitumor immunity primed by Olvi-Vec

❖ **Intraperitoneal (IP) route of delivery is relevant to ovarian cancer**

❖ **Heavily pretreated platinum-resistant/refractory patients who consider clinical trials, standard agents with low response rates, or palliative care**

- **AURELIA study** (limited to platinum-resistant with ≤ 2 prior lines; platinum-refractory patients ineligible; 93% had no prior bevacizumab): ORR: RECIST = 27.3%, CA-125 = 31.8%
 89% of VIRO-15 patients were AURELIA ineligible (refractory or > 2 prior lines)
- **Literature comparisons** in mixed platinum-refractory/resistant patients:
 - ➢ ORR by RECIST at 0 – 19% (mean 9.3%)
 (Griffith 2011; Bruchim 2013; Disis 2019; Konstantinopoulous 2019; Matulonis 2019; Moore 2019; Pujade-Lauraine 2019)
 - ➢ Median PFS ~3 months (1.4 - 6.7 months)
 (Hanker 2012; Bruchim 2013; Ikeda 2013; Davis 2014; Pujade-Lauraine 2014)

Multi-dimensional Mechanism of Action

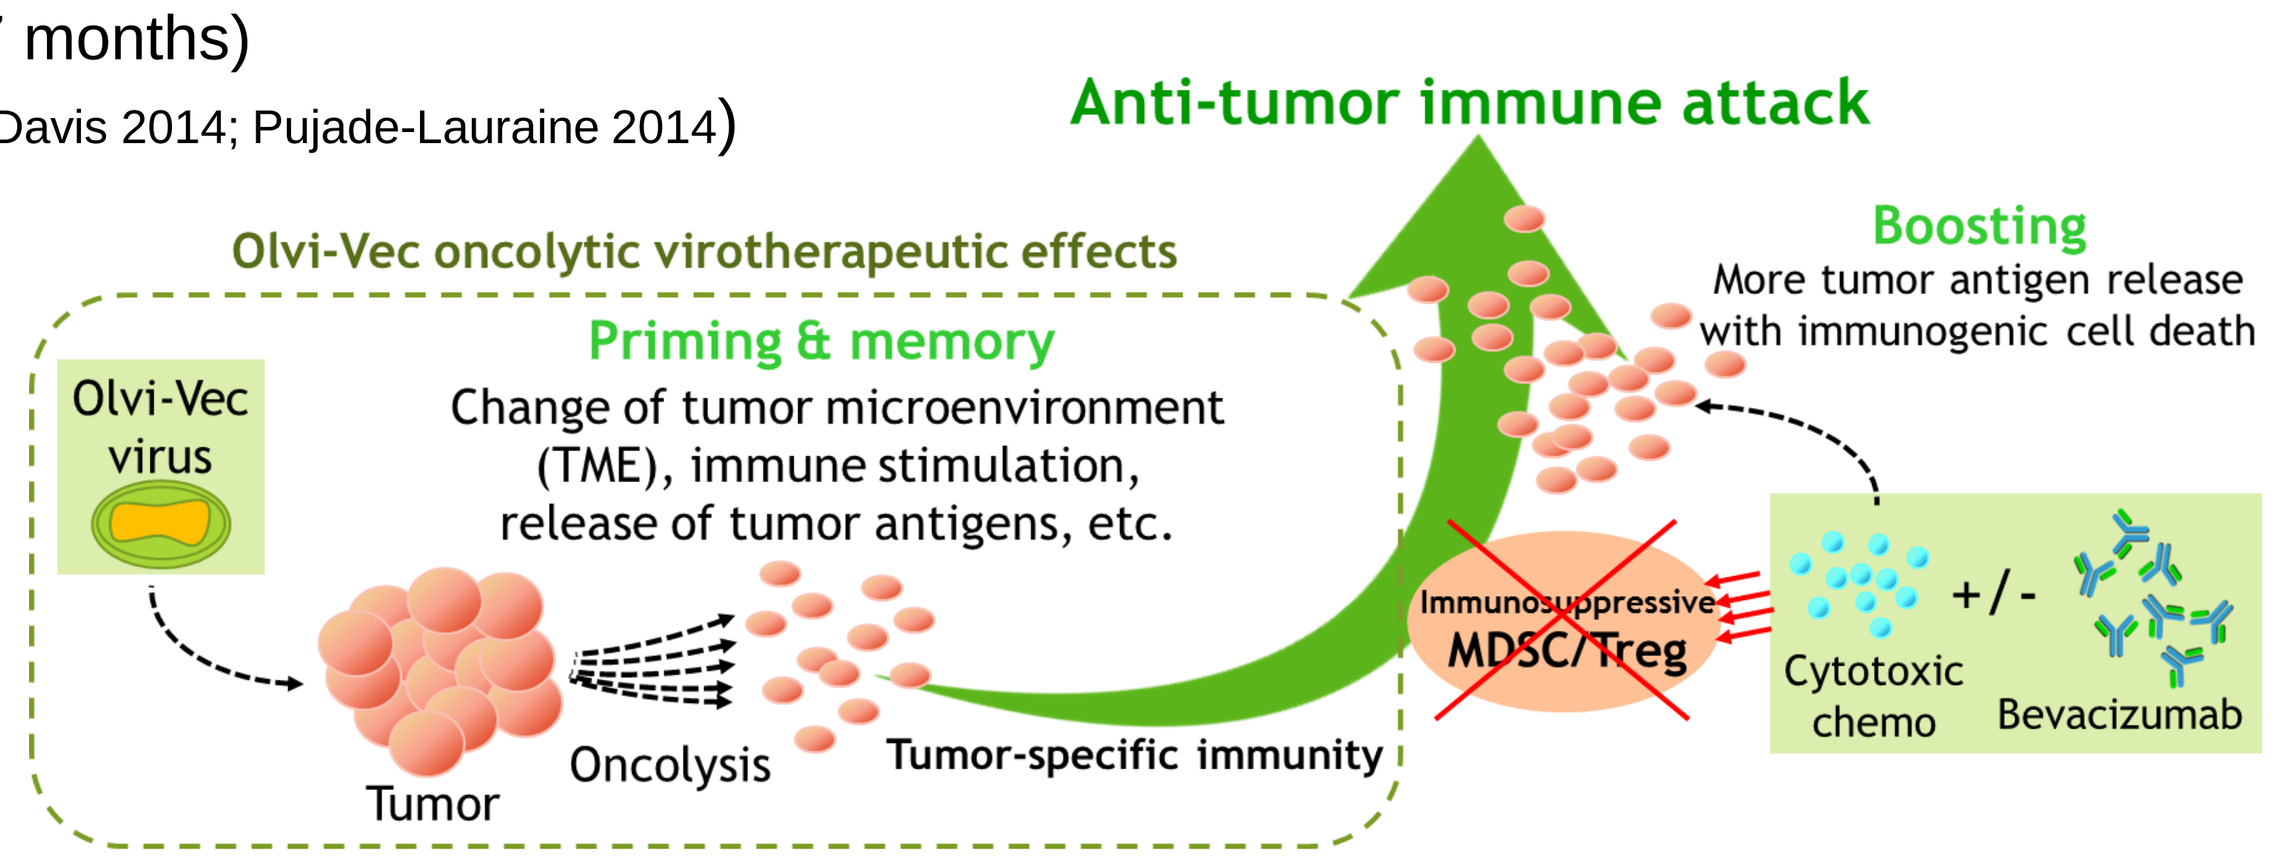
Anti-tumor immune attack
Olvi-Vec oncolytic virotherapeutic effects
Priming & memory
Change of tumor microenvironment (TME), immune stimulation, release of tumor antigens, etc.
Olvi-Vec virus
Tumor
Oncolysis
Tumor-specific immunity
Boosting
More tumor antigen release with immunogenic cell death
Immunosuppressive MDSC/Treg
Cytotoxic chemo
+/-
Bevacizumab

Patients

Baseline Characteristic	Patients (n=27)
Age, median (range)	62 (35-78)
Histology	
High-grade serous	25 (92%)
Intermediate-grade serous	1 (4%)
Mixed	1 (4%)
ECOG performance status	
0	17 (63%)
1	10 (37%)
Prior number of lines, median (range)	4 (2-9)
Prior platinum lines, median (range)	2 (1-5)
Platinum status at enrollment	
Platinum-resistant	13 (48%)
Platinum-refractory	14 (52%)
Prior anti-angiogenic therapy with bevacizumab	
Yes	22 (81%)
No	5 (19%)
Prior PARP inhibitor therapy	
Yes	20 (74%)
No	7 (26%)
Baseline genetic profiles	
Tumor PD-L1 expression	
Positive	1 (4%)
Negative	25 (92%)
Unknown	1 (4%)
BRCA1/2 mutations	
Positive	8 (30%)
Negative	19 (70%)
Microsatellite instability status	
Stable	19 (70%)
Unknown	8 (30%)
Tumor mutational load	
Low	13 (48%)
Intermediate	4 (15%)
Unknown	10 (37%)
Response & PFS from last prior line before enrollment into VIRO-15 trial	
ORR by RECIST	4/27 (15%)
ORR by CA-125	5/24 (21%)
PFS (mos), median (95% CI)	4.5 (2.9,5.8)
PFS-6-month	29%

Efficacy Results

Overall Response Rate (ORR), Progression-Free Survival (PFS), and Overall Survival (OS)

All patients had documented progressive disease at enrollment into VIRO-15 trial. Results are in eligible for evaluation patients.

	ORR by RECIST1.1	Duration of Response	ORR by CA-125	Median PFS	Median OS
All patients (n= 27) (95% CI)	**54%** (13/24) (33, 74)	**7.6 mos** (3.7, 9.6)	**85%** (22/26) (65, 96)	**11.0 mos** (6.7, 13.0)	**15.7 mos** (12.3, 23.5)
Platinum-resistant (n=13) (95% CI)	**60%** (6/10) (26, 88)	**7.6 mos** (3.7, NA)	**85%** (11/13) (55, 98)	**11.0 mos** (5.2, NA)	**17.4 mos** (6.9, 23.5)
Platinum-refractory (n=14) (95% CI)	**50%** (7/14) (23, 77)	**8.0 mos** (3.7, NA)	**85%** (11/13) (55, 98)	**10.8 mos** (4.3, 13.2)	**15.2 mos** (10.8, 33.6)

- Disease Control Rate (CR + PR + SD): 86% (24/27)
- 4 patients had 100% reduction of target lesions including platinum-refractory patients
- 96% (25/26) of patients achieved decrease of CA-125 tumor biomarker
- All 11 patients with > 90% decrease of CA-125 achieved RECIST response
- RECIST responses correlate to CA-125 responses (p = 0.007)

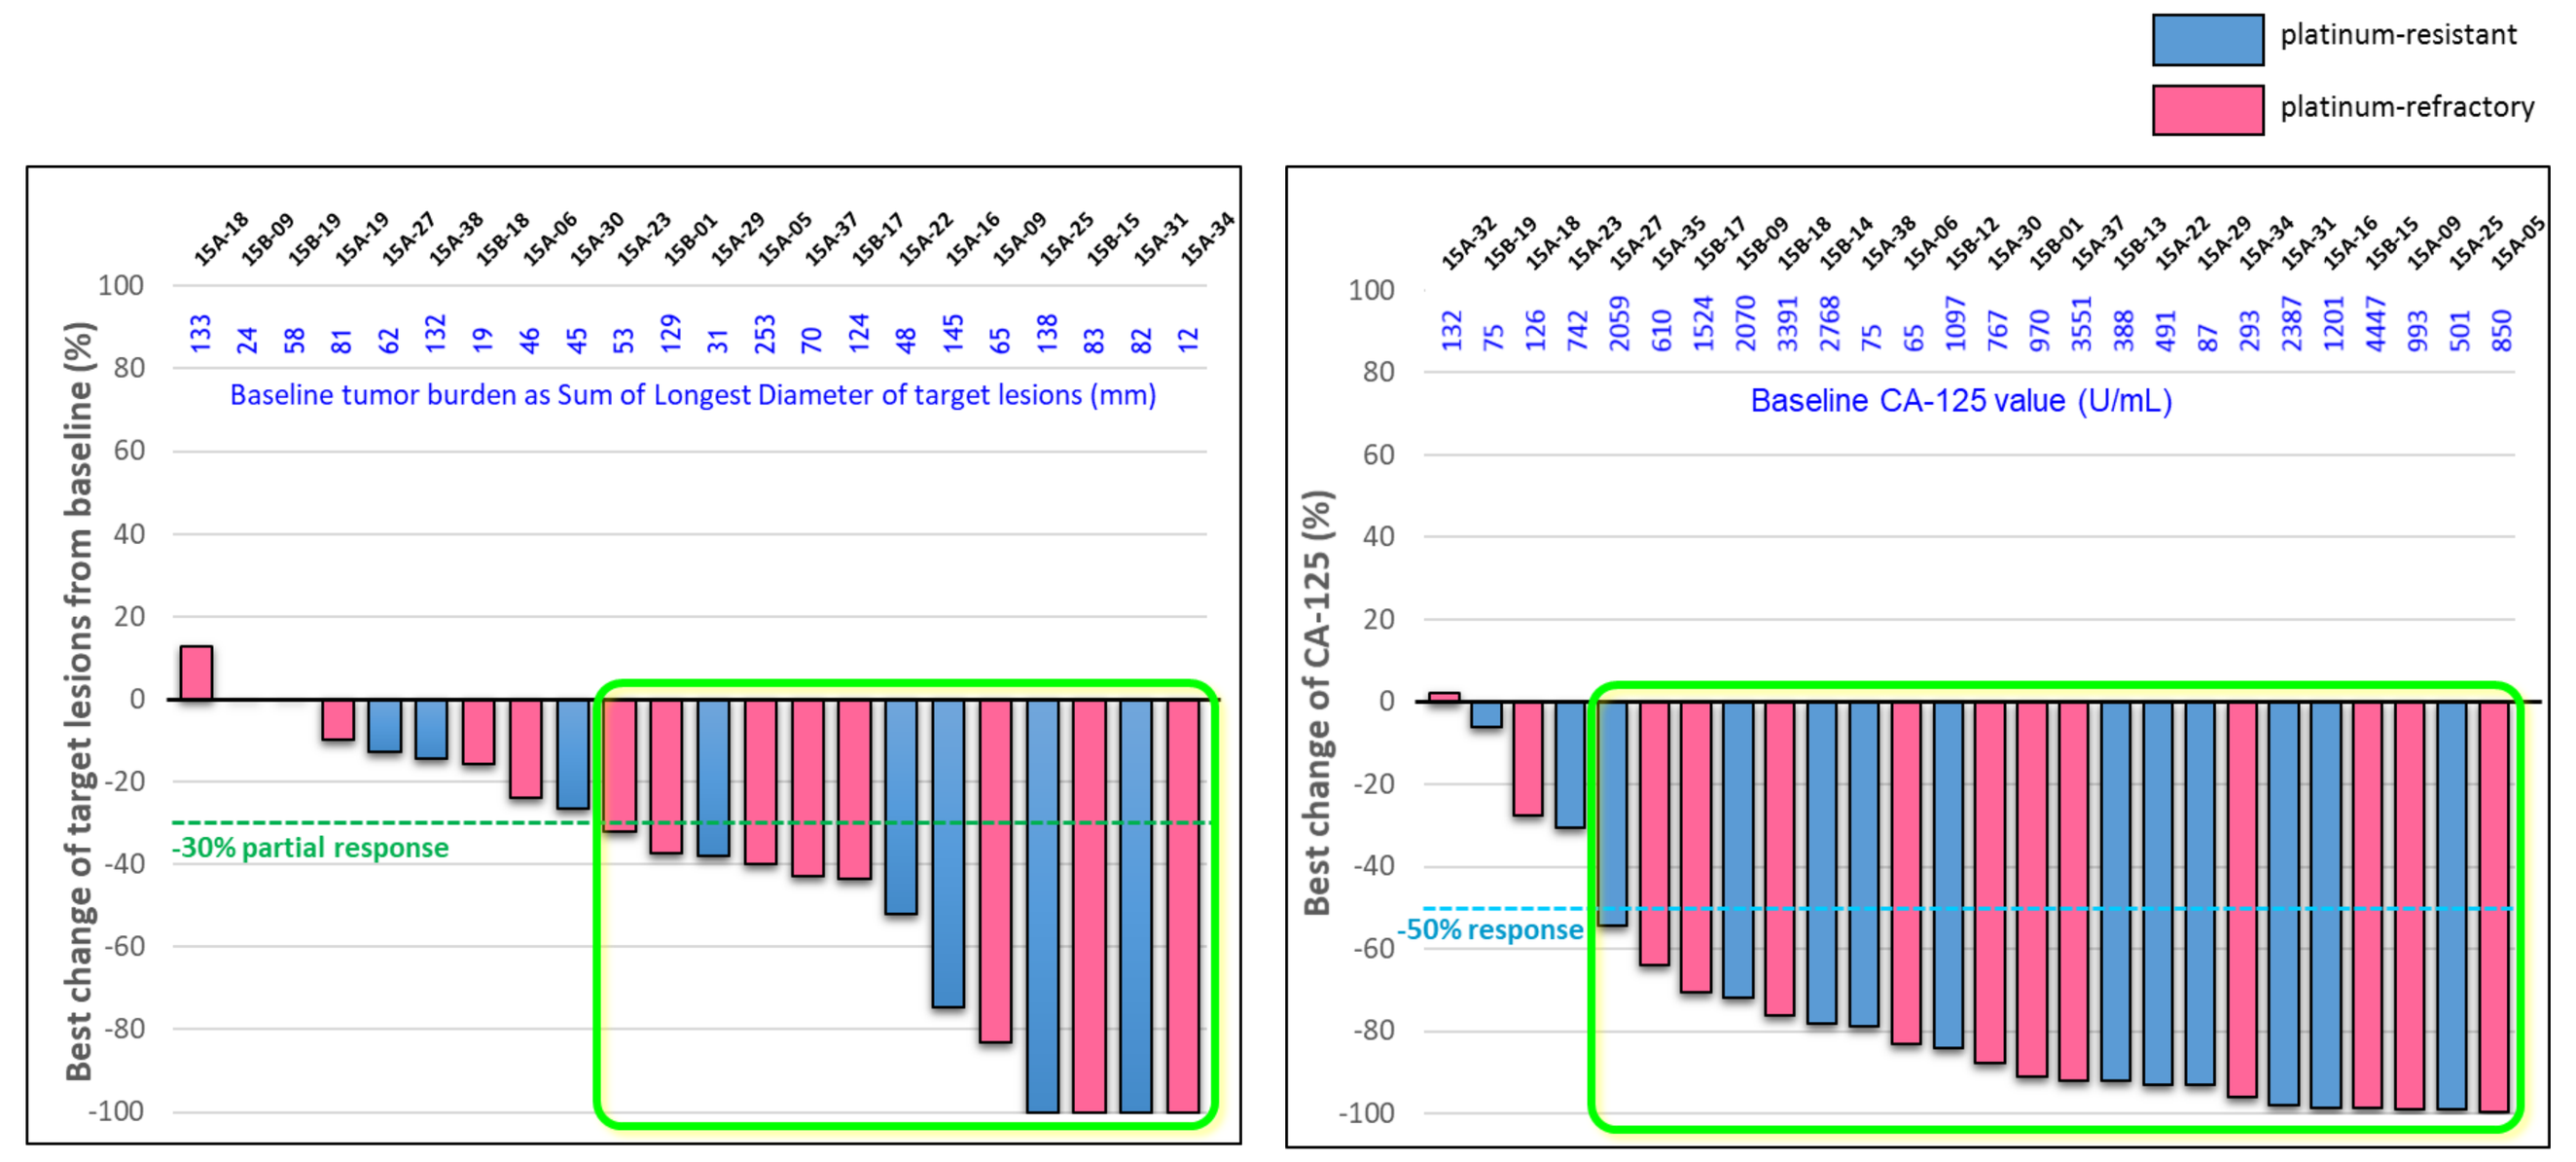
platinum-resistant
platinum-refractory
Baseline tumor burden as Sum of Longest Diameter of target lesions (mm)
Best change of target lesions from baseline (%)
-30% partial response
Baseline CA-125 value (U/mL)
Best change of CA-125 (%)
-50% response

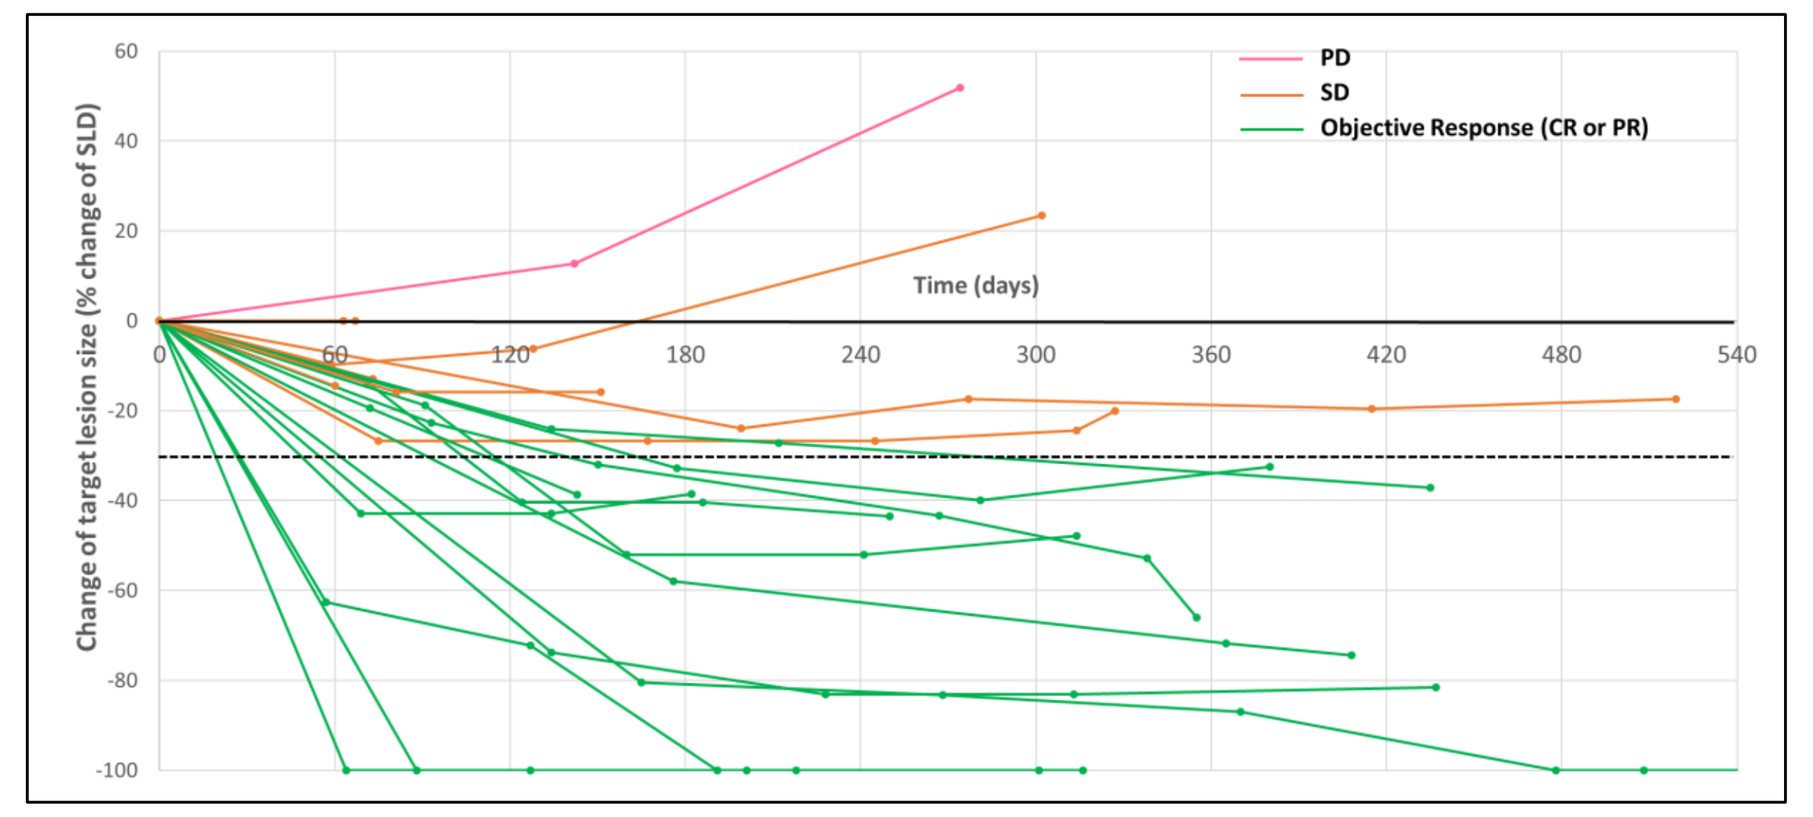
PD
SD
Objective Response (CR or PR)
Time (days)
Change of target lesion size (% change of SLD)

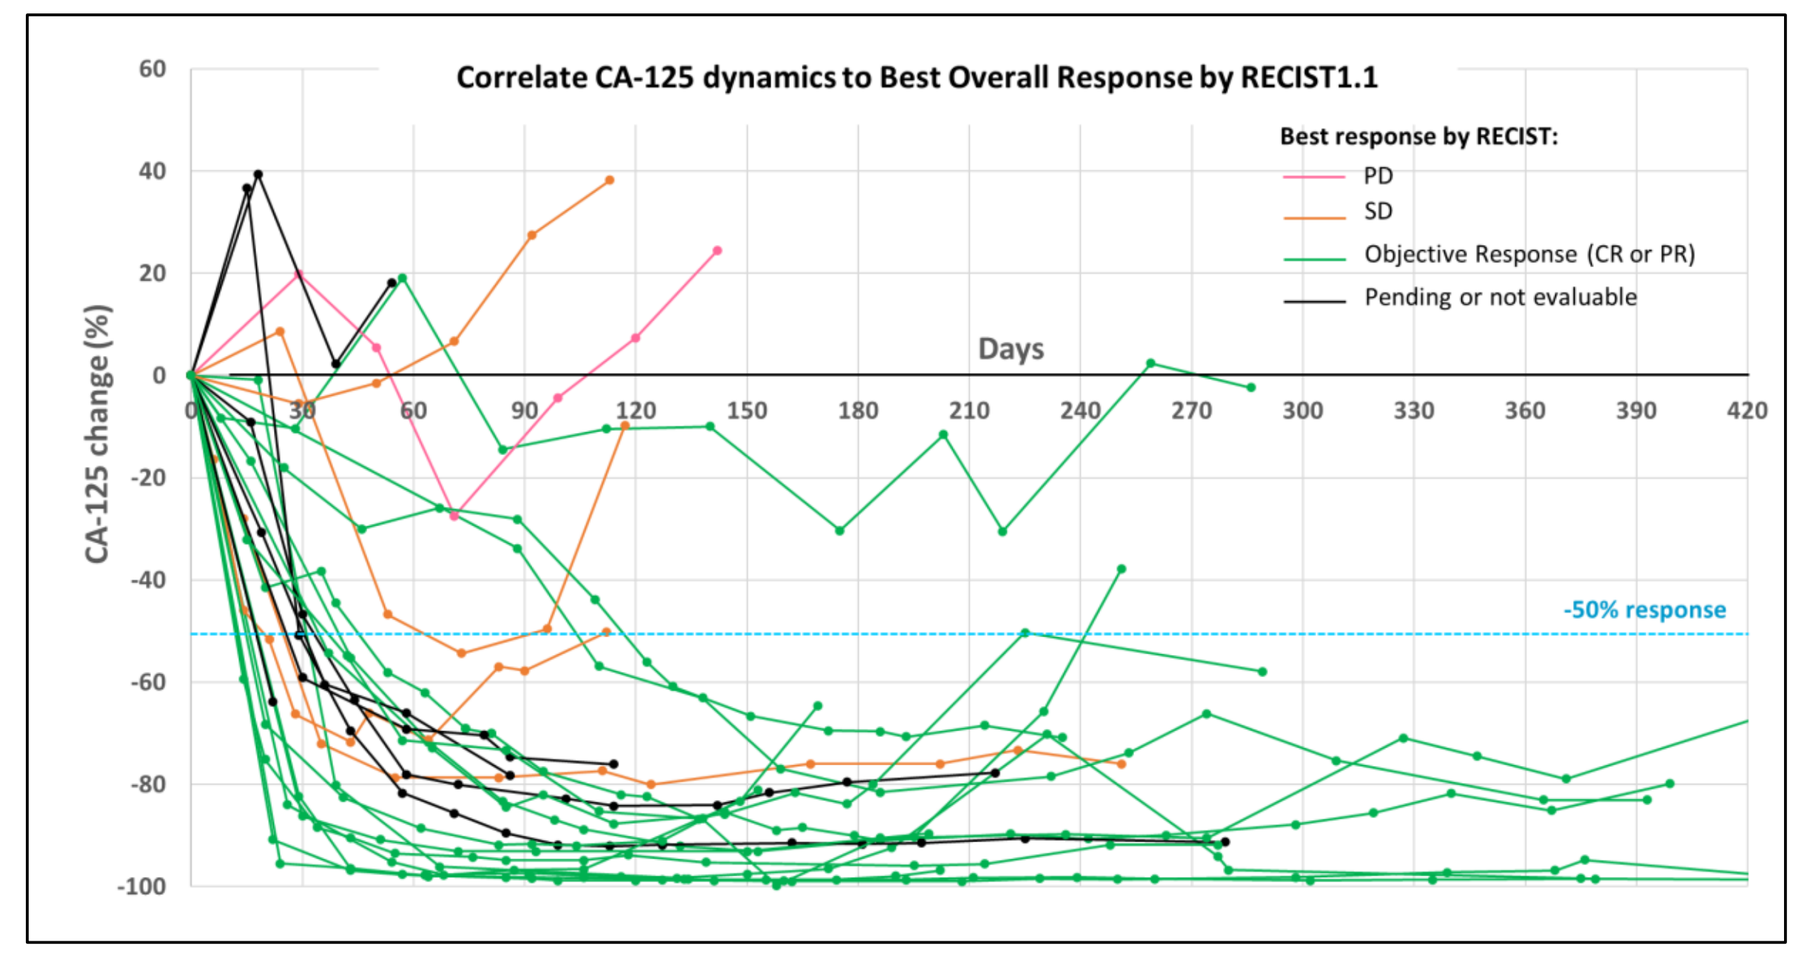
Correlate CA-125 dynamics to Best Overall Response by RECIST1.1
Best response by RECIST:
PD
SD
Objective Response (CR or PR)
Pending or not evaluable
Days
CA-125 change (%)
-50% response

Safety Results (Olvi-Vec)

Grades 1 & 2 Adverse Events (in ≥ 20% patients)	
Pyrexia	16 (59%)
Nausea	13 (48%)
Abdominal distension	12 (44%)
Abdominal pain	12 (44%)
Chills	10 (37%)
Fatigue	9 (33%)
Vomiting	7 (26%)
Grade 3 Adverse Events (in ≥ 2 patients)	
Abdominal pain	2 (7%)
Hypophosphatemia	2 (7%)
No Grade 4 AEs	

- Safety results consistent with previous Phase 1b trial
- Prophylactic hydrations helpful following IP Olvi-Vec

Conclusions

❖ **Phase 2 VIRO-15 trial meets the co-Primary Endpoints of ORR & PFS after median follow-up of 36.0 months in *heavily pretreated* patients with PRROC**

All PRROC patients (median 4 prior lines):

- **54% response by RECIST1.1**
- **7.6 months median Duration of Response per RECIST1.1**
- **85% response by CA-125**
- **11.0 months median PFS, significantly greater than last treatment (4.5 months)**
- **77% PFS-6-month (vs. 29% from last prior line in the same patients)**
- **15.7 months median overall survival**

Sub-population of platinum-refractory patients (median 4 prior lines):

- **50% response by RECIST1.1**
- **8.0 months median Duration of Response per RECIST1.1**
- **85% response by CA-125**
- **10.8 months median PFS, significantly greater than prior treatment in the same patients (4.6 months)**
- **15.2 months median overall survival**

❖ **Translational data indicate that Olvi-Vec increases intratumoral (particularly intraepithelial) CD8+ TILs, generates systemic tumor-specific T-cells response, and upregulates intratumoral STAT1 gene expression of the IFN pathways to favorably affect the tumor microenvironment, and clinically overcome platinum resistance with substantial response to subsequent platinum doublet therapy.**

❖ **A registration trial of Olvi-Vec-primed immunochemotherapy is under review for 2021.**

Objectives & Methods

VIRO-15:
Multi-center, open-label, non-randomized Phase 2 study of IP Olvi-Vec followed by IV carboplatin doublet ± bevacizumab in PRROC patients

- ❖ **Primary Endpoints:** ORR by RECIST1.1 & by CA-125; PFS
- ❖ **Secondary Endpoints:** Adverse events; Duration of Response, OS
- ❖ **Translational Endpoints:** TILs; gene expression in paired tumor biopsies; tumor-specific T-cell response in blood

Inclusion Criteria

- Platinum-resistant or -refractory disease
- Epithelial ovarian cancer with high grade serous, endometrioid, clear cell
- $\geq$ 2 prior lines of therapy
- Progressed on last line of therapy
- ECOG PS: 0 or 1

Exclusion Criteria

- No visible intraperitoneal disease
- Rapidly progressive pleural effusions or ascites
- Prognostic Nutritional Index (PNI) < 40 (suggested)

Translational Analyses

Olvi-Vec-induced favorable systemic & intratumoral immune activations

- Large intra-tumoral (particularly intraepithelial) infiltration of CD4+ & CD8+ T-cells shown by multiplex IHC (photos and histograms below) .
- Tumor gene expression was analyzed in paired (before and after virotherapy) tumor biopsies, with STAT1 up-regulation having the most significant p-value (p=0.008) among 770 analyzed genes (NanoString RNA profiling).

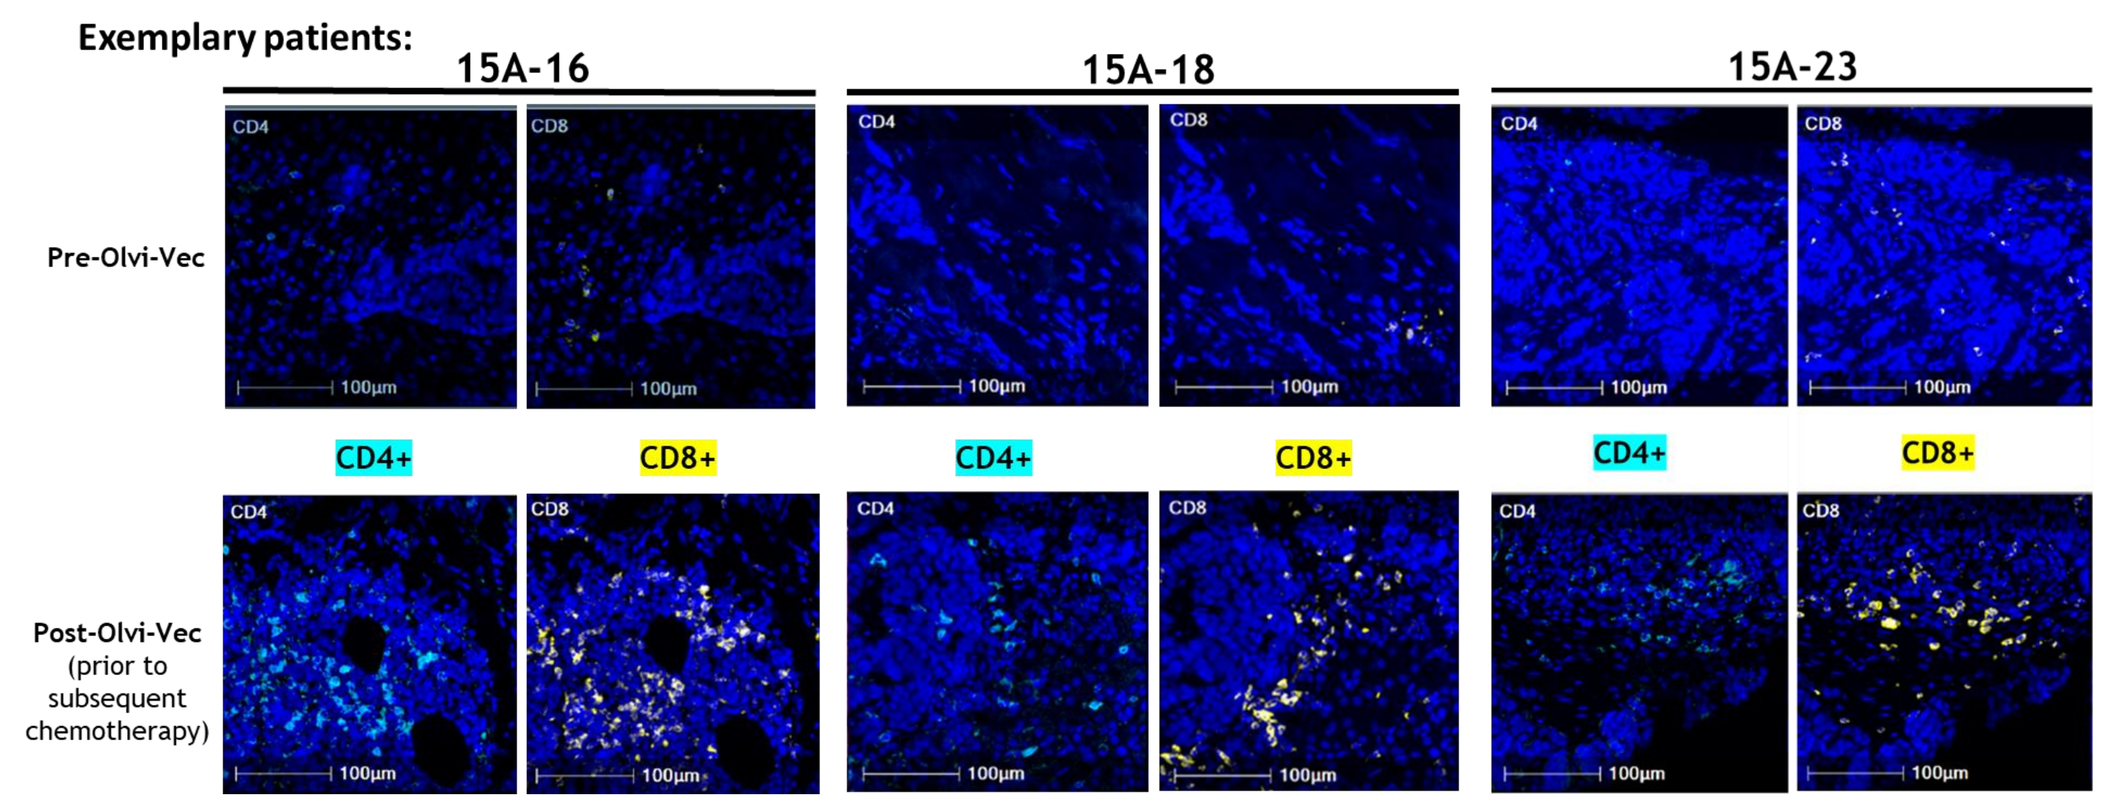
Exemplary patients:
15A-16
15A-18
15A-23
Pre-Olvi-Vec
Post-Olvi-Vec (prior to subsequent chemotherapy)
CD4+
CD8+

CD8+ infiltrating T Cells before (A) and after (B) IP Olvi-Vec virotherapy

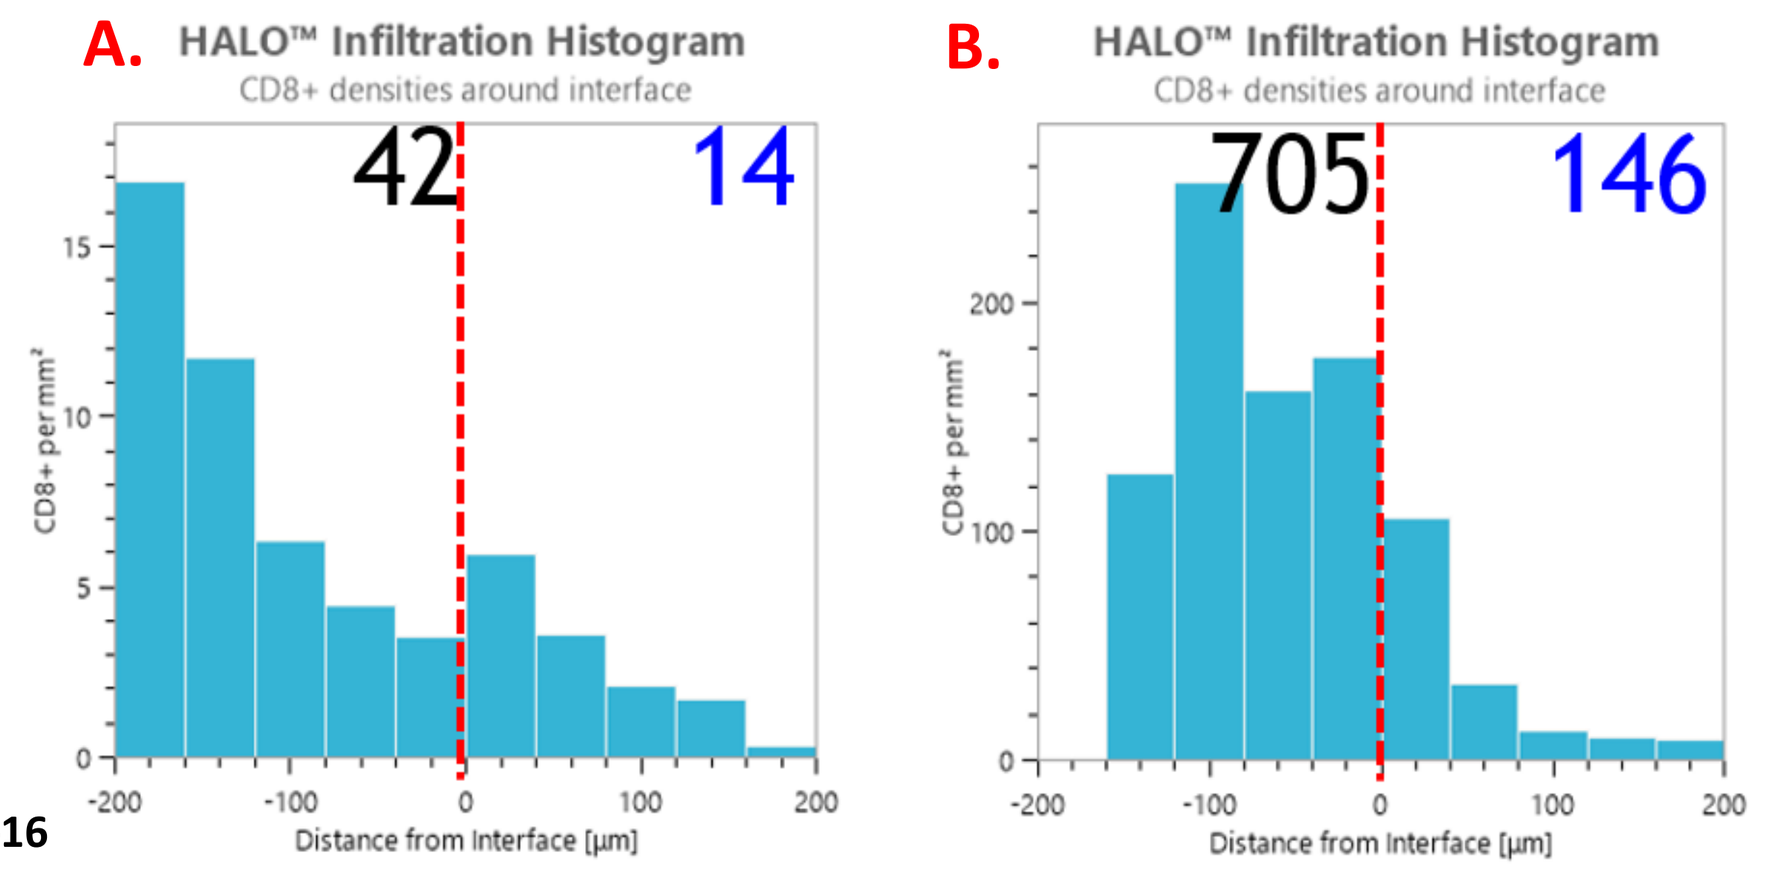
A. HALO™ Infiltration Histogram
CD8+ densities around interface
42
14
B. HALO™ Infiltration Histogram
CD8+ densities around interface
705
146
CD8+ per mm²
Distance from Interface [µm]

Exemplary Patient: 15A-16

References:

- Au *et al*., J Pathol Clin Res. 2016;2(4):259–270
- Binz et al., Mol Ther Oncolytics. 2017; 6: 10–21
- Bruchim *et al*., Eur J Obs Gyn Rep Biol. 2013;166(1):94-8
- Davis *et al*., Gynecol Oncol. 2014;133(3):624-31
- Disis *et al*., JAMA Oncol. 2019;5(3):393-401
- Emens & Middleton, Cancer Immunol Res. 2015;3(5):436-43
- Griffith *et al*., Int J Gynecol Cancer. 2011;21(1):58-65
- Hanker *et al*., Ann Oncol. 2012;23(10):2605-12
- Ikeda *et al*., Int J Gynecol Cancer. 2013;23(2):355-60
- Kilinc *et al*., J Transl Med 2016;14:340
- Konstantinopoulos *et al*., JAMA Oncol. 2019;5(8):1141–9
- Matulonis *et al*., Ann Oncol. 2019;30(7):1080-7
- Moore *et al*., Lancet Oncol. 2019;20(5):636-8
- Pujade-Lauraine *et al*., J Clin Oncol. 2014;32(13):1302-8
- Pujade-Lauraine *et al*., SGO 2019 Annual Meeting
- Song *et al*., Mol Ther. 2007;15(8):1558-63
- Von Hoff *et al*., J. Clin. Oncol. 2010;28(33):4877-83
- Wang *et al*., Cell. 2016;165(5):1092-105
- Worschech *et al*., Cancer I. Immunother. 2009;58(9): 1355–62
- Zhang *et al*., Mol Genet Genomics. 2009;282(4):417-35

Acknowledgement:
All staff at Genelux Corporation for support;
Study sponsored by Genelux Corporation

Corresponding author:
robhollowaymd@gmail.com

VIRTUAL 2020 ESMO congress
European Society for Medical Oncology (ESMO) Virtual Congress 2020; September 19-21, 2020.

Protocol #: GL-ONC1-015; ClinicalTrials.gov Identifier: NCT02759588; Copyright © 2020 Genelux Corp. All rights reserved.